Investigation of the Reinforcing and Discriminative Properties of Plant-Derived, Highly Purified Cannabidiol (CBD) in Rats and Monkeys

Roy Gray[1], David Heal[2], David Maguire[3], Lisa Gerak[3], Marty Javors[3], Sharon Smith[2], Charles France[3]

[1]GW Research Ltd., Cambridge, UK;.[2]RenaSci Ltd., Nottingham, UK; [3]University of Texas Health, San Antonio, TX, USA.

Poster M233

SUMMARY

- Plant-derived, highly purified CBD (Epidiolex® 100 mg/mL oral solution) is approved in the US for the treatment of seizures associated with Lennox-Gastaut syndrome or Dravet syndrome in patients ≥2 years of age.
- Studies were conducted in accordance with FDA Guidance (Assessment of Abuse Potential of Drugs (2017)) in support of NDA application and represent a component of the 8-factor analysis (8FA) for determination of drug scheduling in the USA.
- Accordingly, the discriminative properties of CBD were assessed in rats trained to midazolam and reinforcing properties were assessed using the *i.v.* self-administration procedure in rats trained to heroin and rhesus monkeys trained to midazolam.
- CBD, at doses that produced systemic exposure equal to and greater than those observed in humans, was not a strong positive enforcer and did not produce an interoceptive effect similar to midazolam.
- These data contributed to the 8FA for Epidiolex (a specific formulation of plant-derived, highly purified CBD) and its Schedule V Controlled Drug classification in the USA.
- These data strongly suggest that CBD possesses negligible potential for human abuse.

RESULTS

Drug discrimination in midazolam-trained rat

A

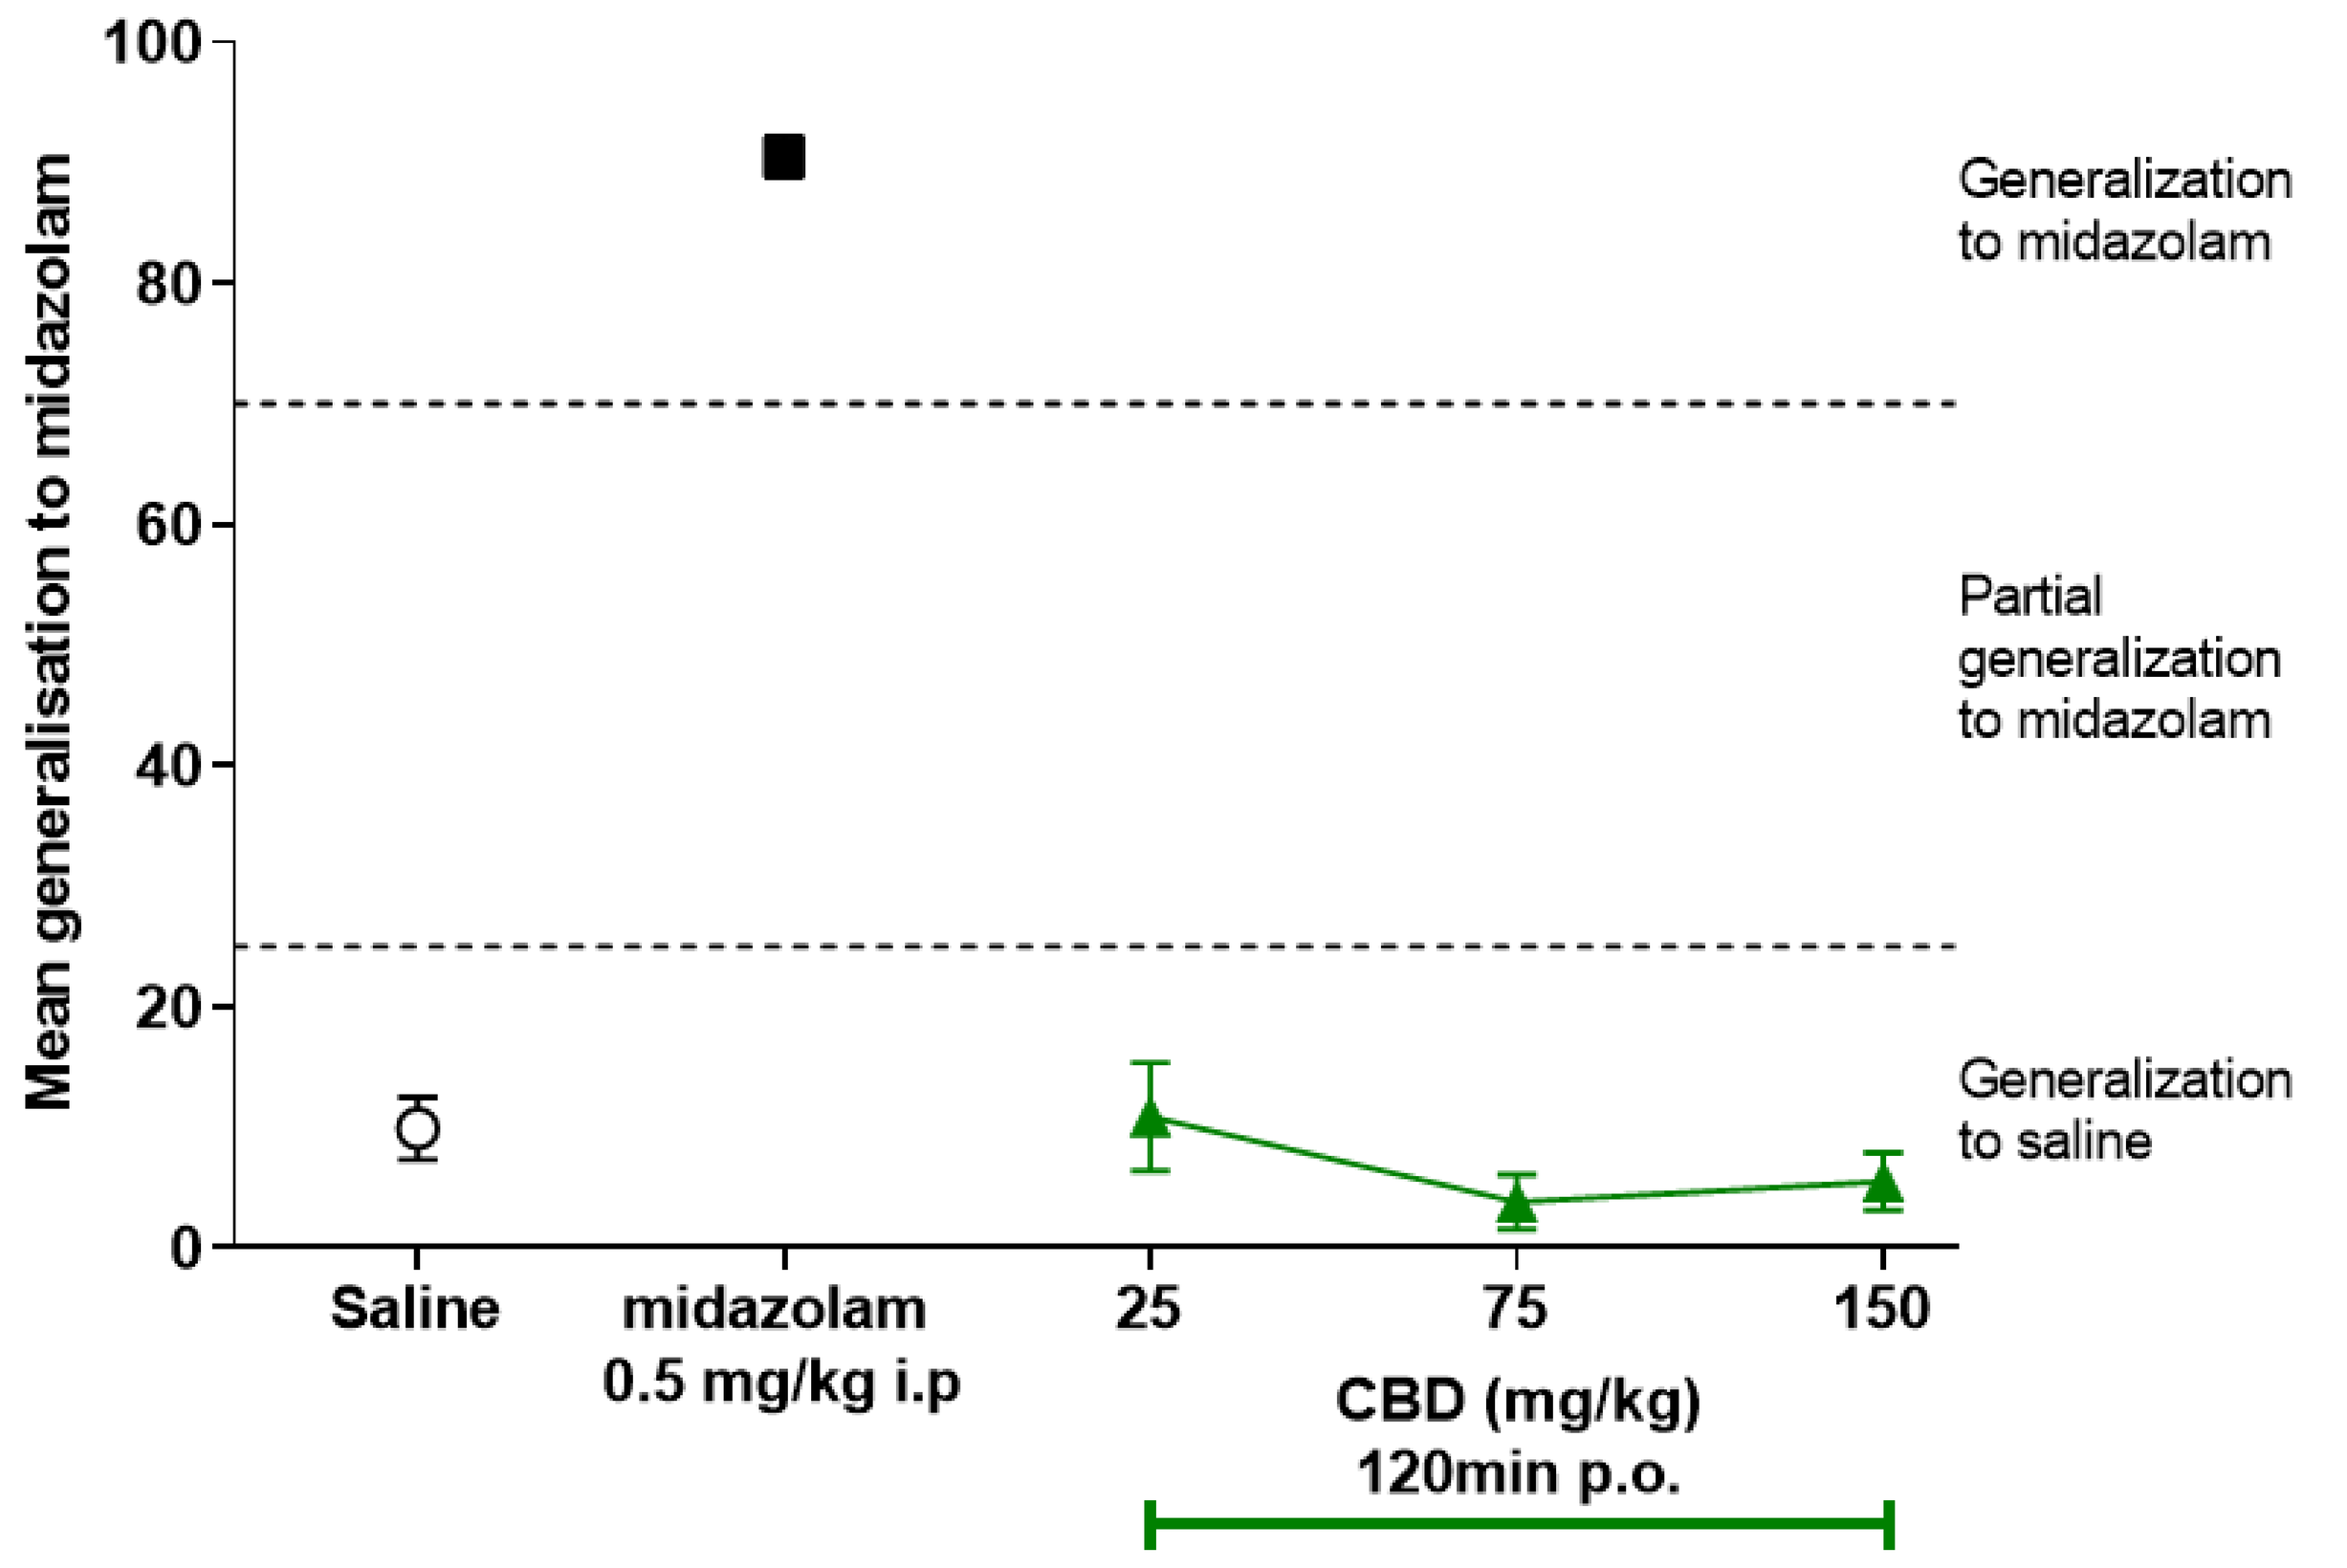
Mean generalisation to midazolam
100
80
60
40
20
0
Generalization to midazolam
Partial generalization to midazolam
Generalization to saline
Saline
midazolam 0.5 mg/kg i.p
25
75
150
CBD (mg/kg) 120min p.o.

B

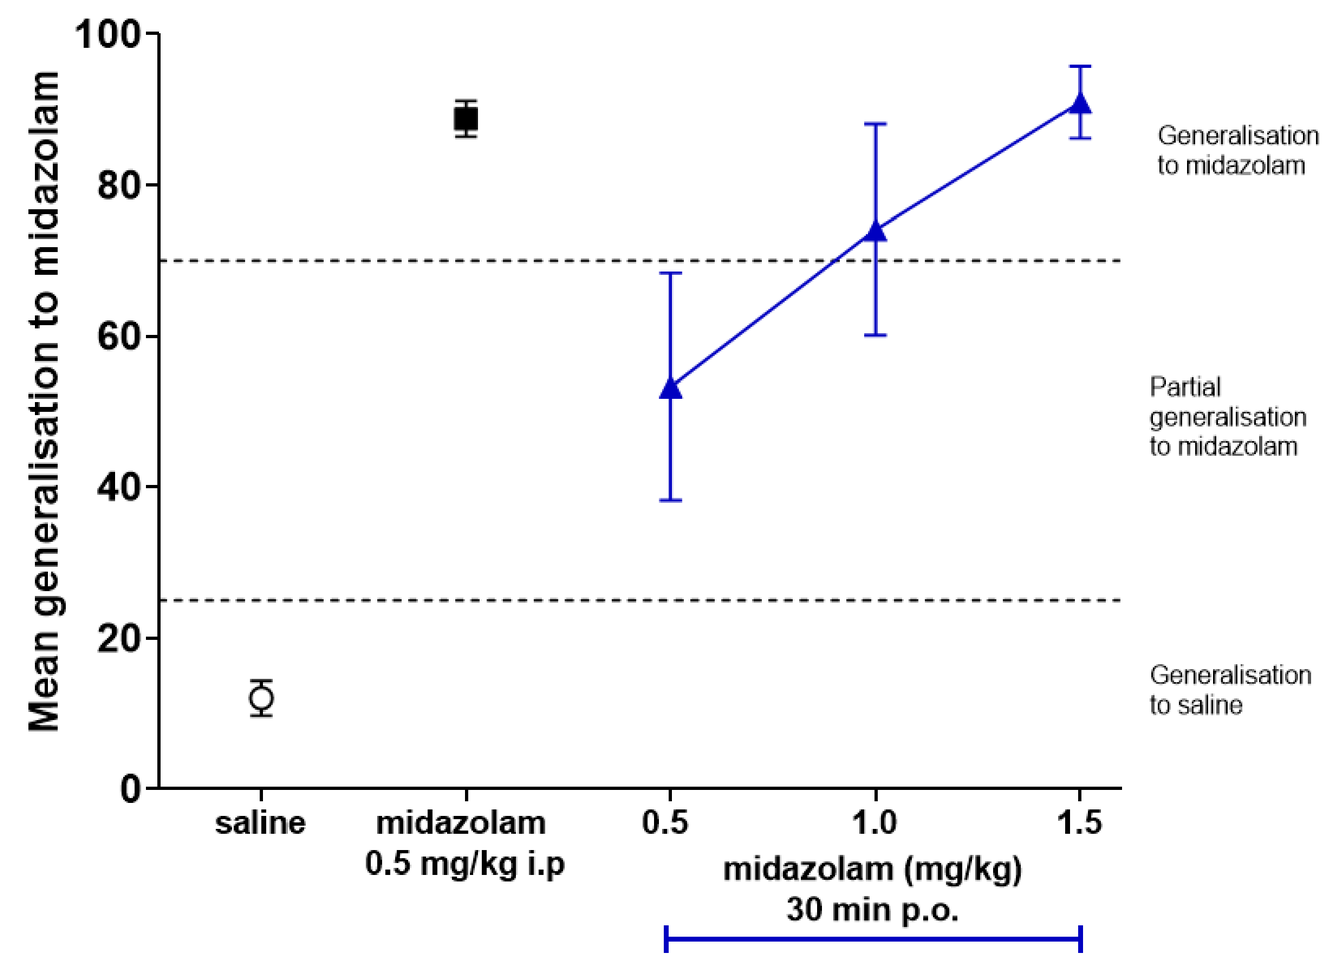
Mean generalisation to midazolam
Generalisation to midazolam
Partial generalisation to midazolam
Generalisation to saline
saline
midazolam 0.5 mg/kg i.p
0.5
1.0
1.5
midazolam (mg/kg) 30 min p.o.

C

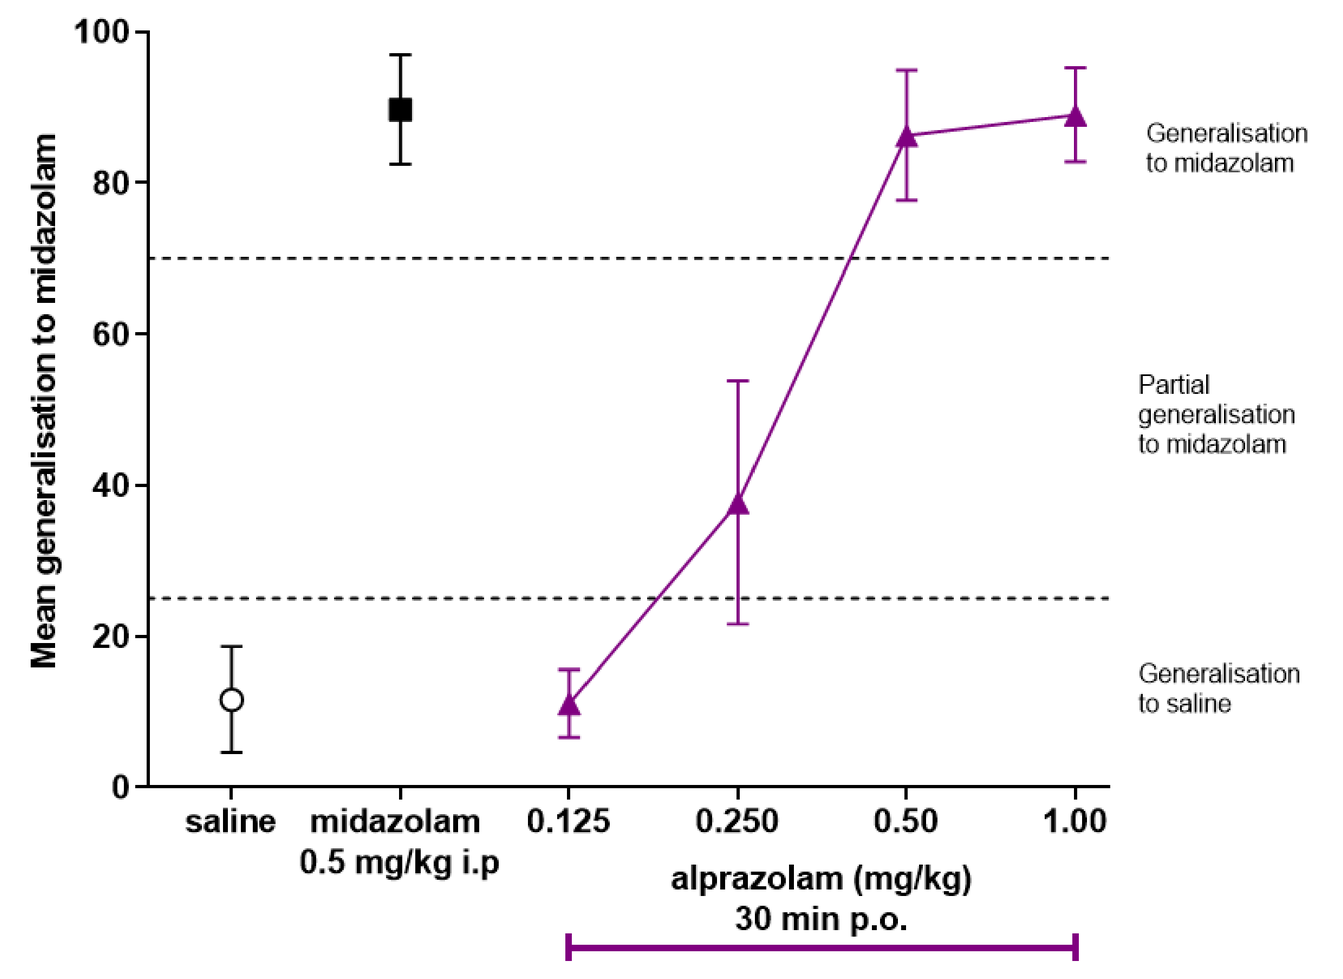
Mean generalisation to midazolam
Generalisation to midazolam
Partial generalisation to midazolam
Generalisation to saline
saline
midazolam 0.5 mg/kg i.p
0.125
0.250
0.50
1.00
alprazolam (mg/kg) 30 min p.o.

Effects of orally administered (A) CBD; (n=6), (B) midazolam (n=6-8) and (C) alprazolam (n=6-13) in rats trained to discriminate midazolam (0.5 mg/kg i.p.) from saline determined 120 min (CBD) or 30min (midazolam & alprazolam) after dosing. Data shown as mean % generalisation to midazolam ± SEM. Saline and midazolam (0.5 mg/kg i.p.) data represent the mean of the four test sessions preceding the first oral dose of test compound.

- In midazolam-cued drug-discrimination, all orally administered CBD doses (20, 75, 150 mg/kg) generalized to the saline cue (Fig A).
- Orally administered midazolam (Fig. B) and alprazolam (Fig. C) dose-dependently generalised to the discriminative cue elicited by intraperitoneally injected midazolam (0.5 mg/kg).

Evaluation of reinforcing effects of CBD in the heroin-trained rat

A

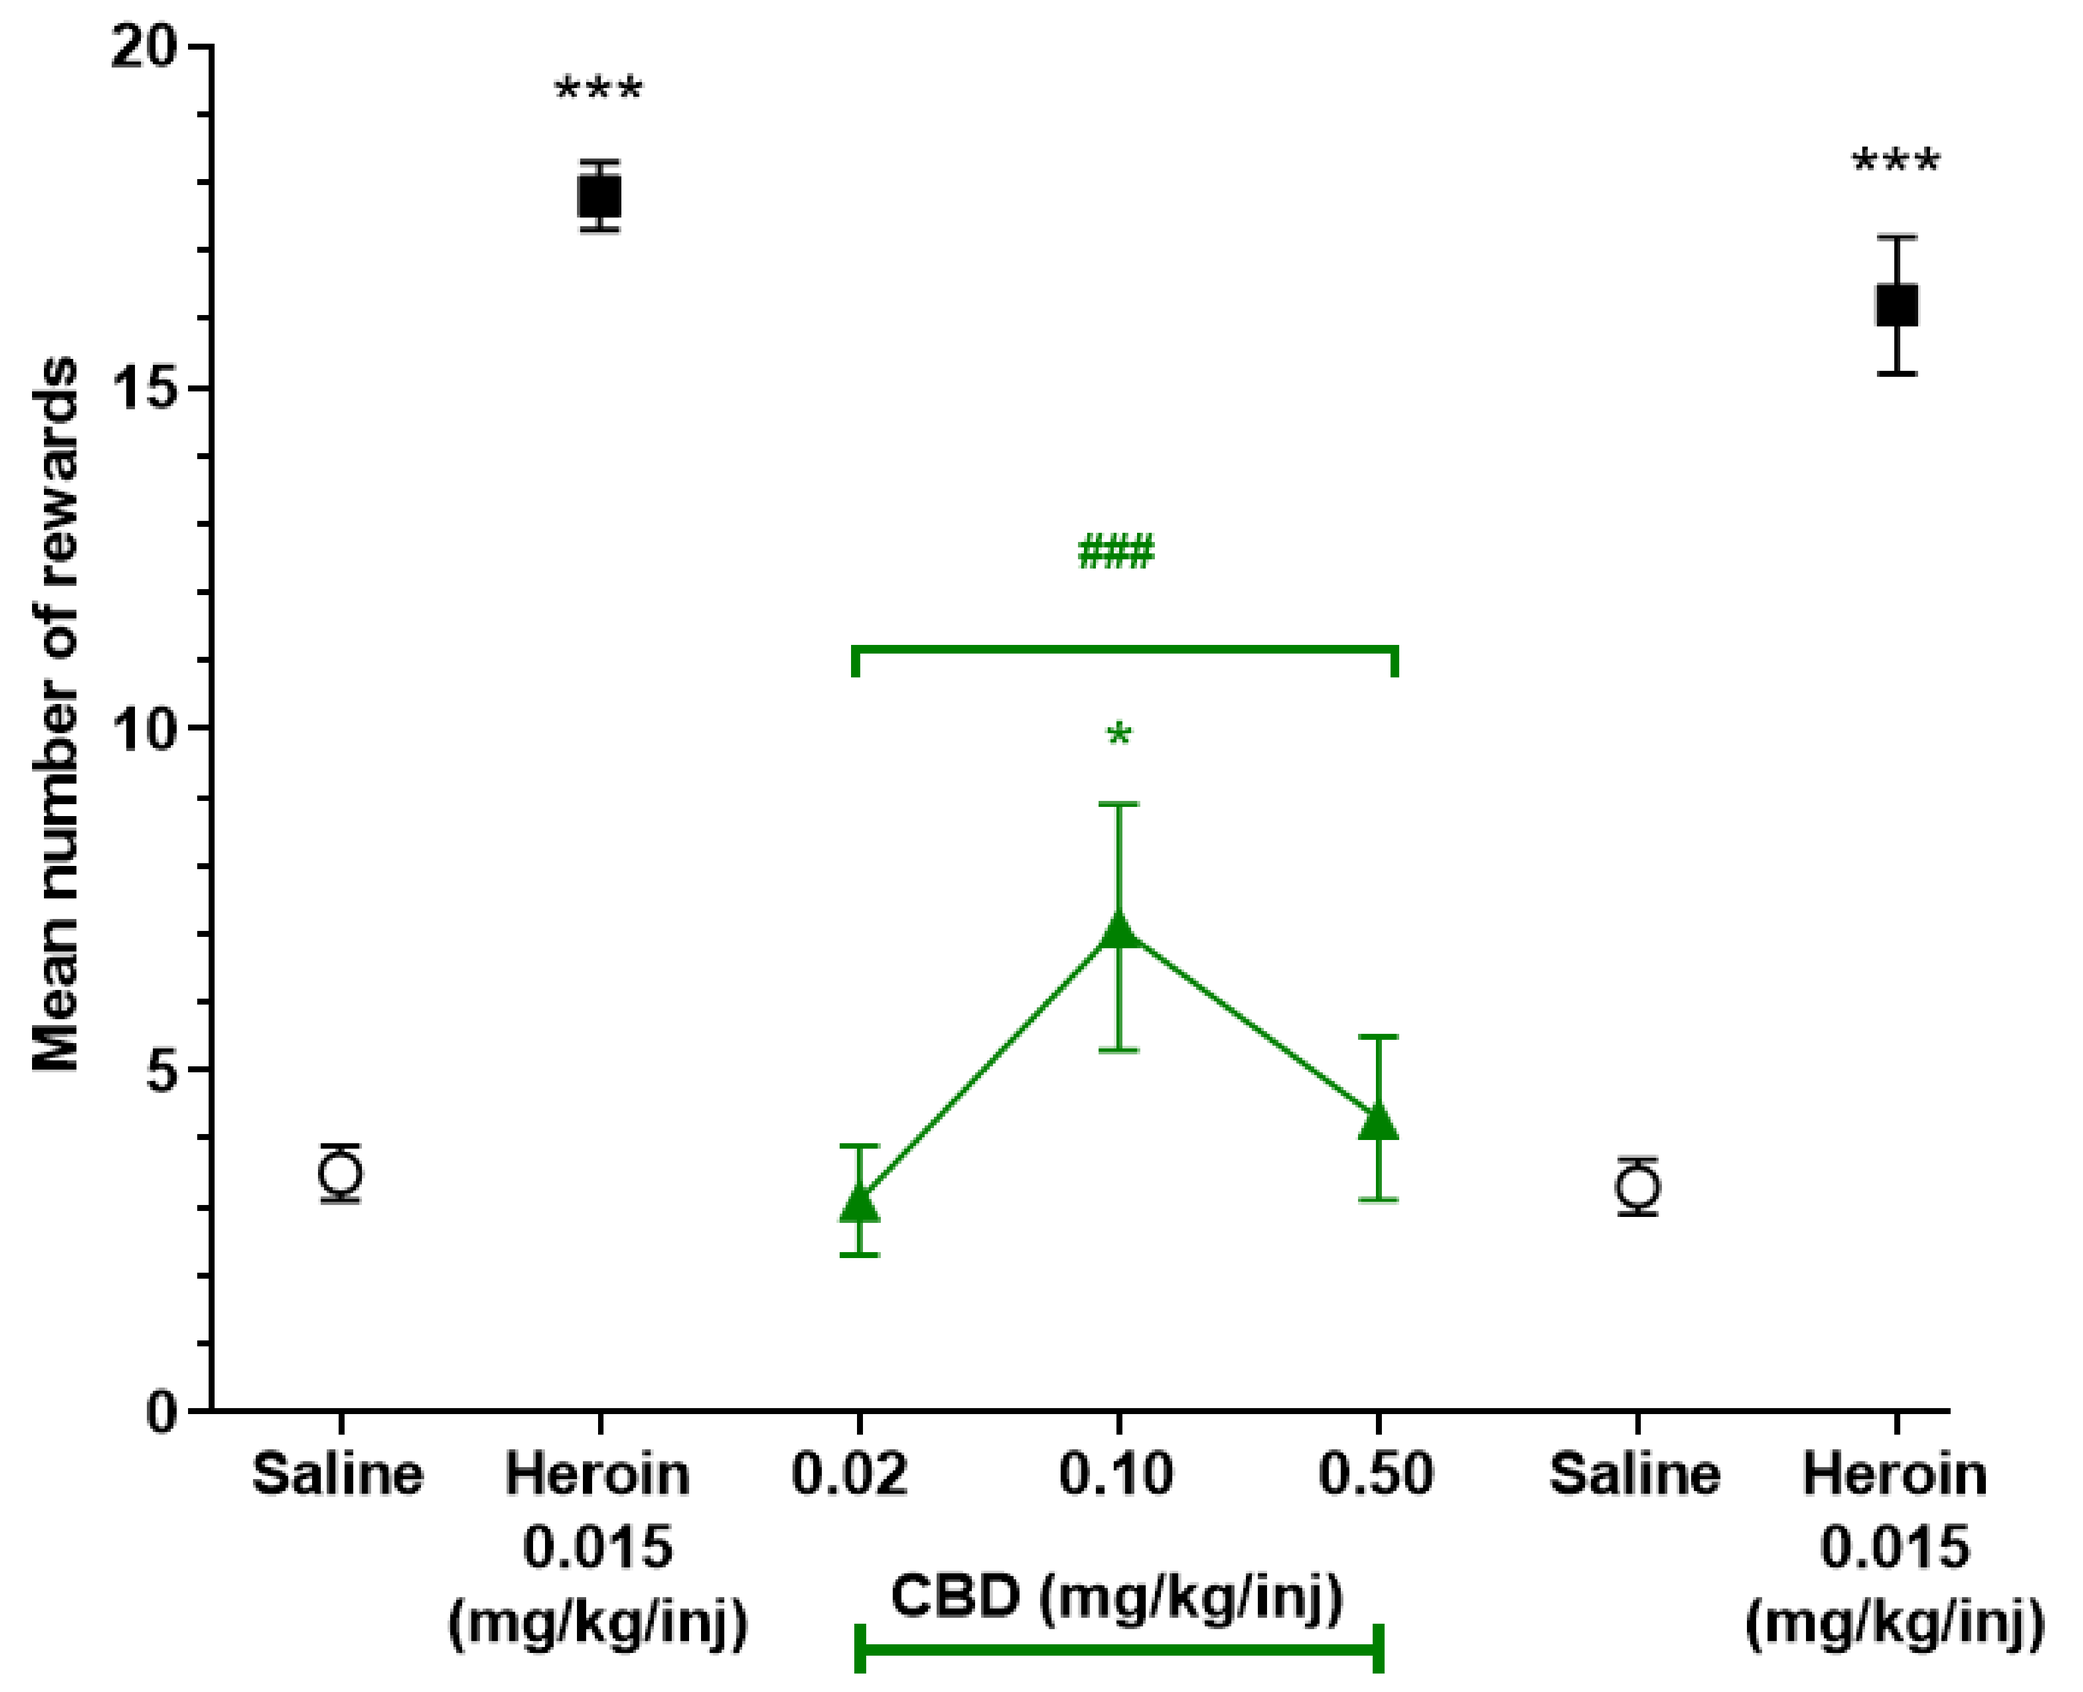
Mean number of rewards
20
15
10
5
0

###
*

Saline
Heroin 0.015 (mg/kg/inj)
0.02
0.10
0.50
CBD (mg/kg/inj)
Saline
Heroin 0.015 (mg/kg/inj)

B

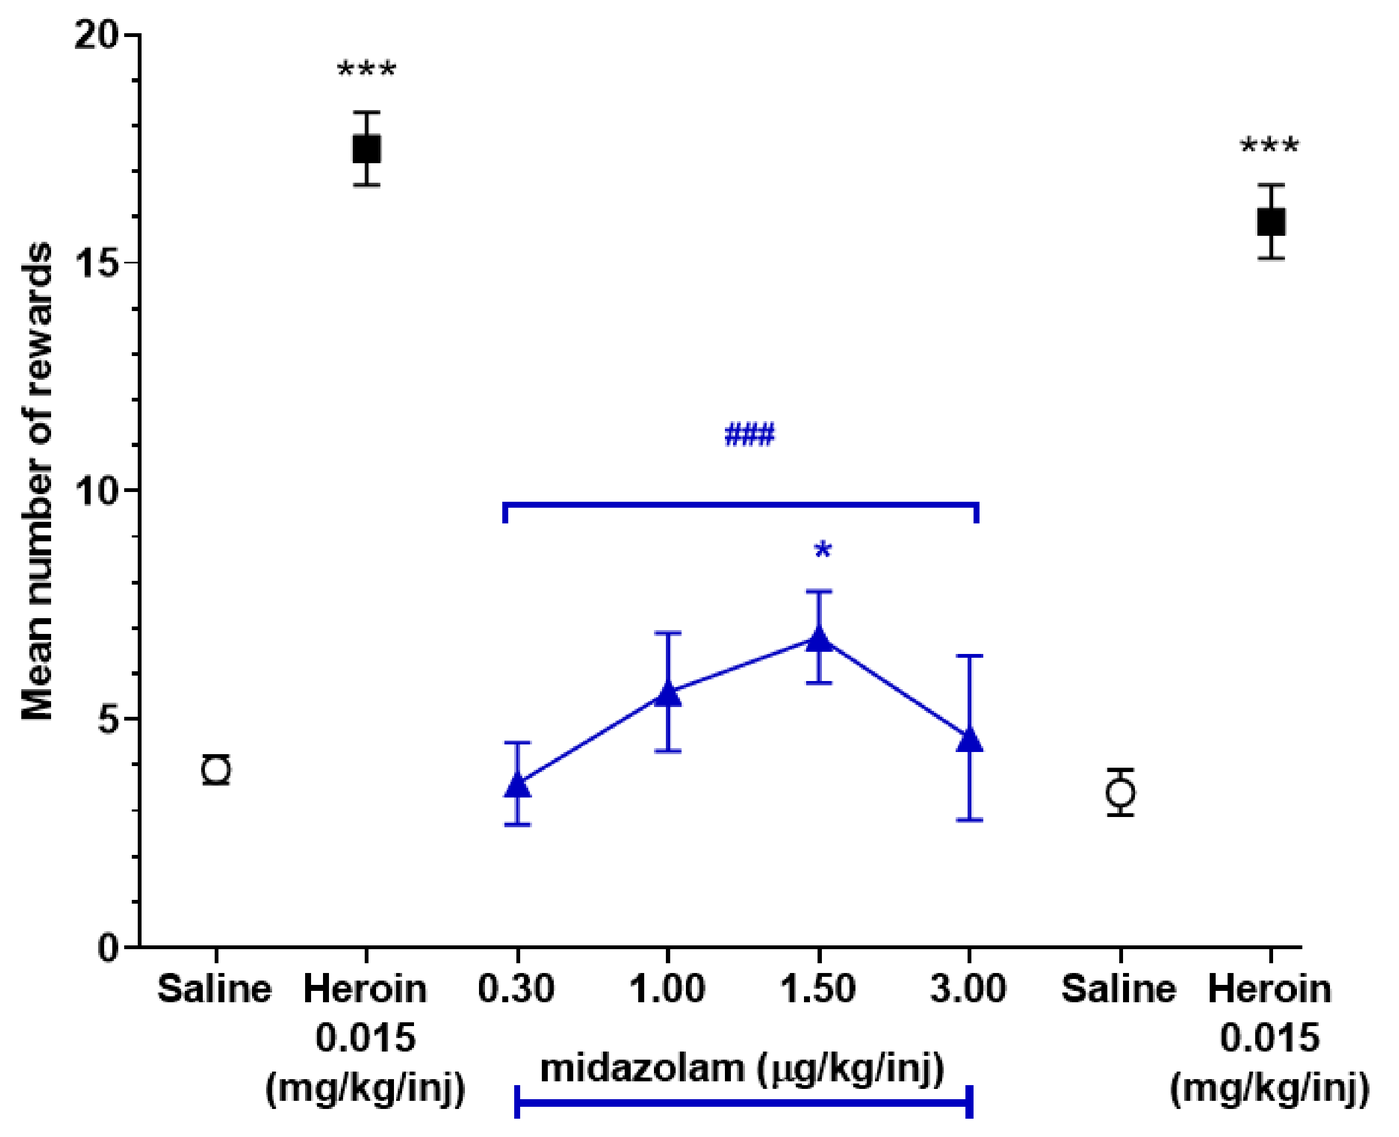
Mean number of rewards

###
*

Saline
Heroin 0.015 (mg/kg/inj)
0.30
1.00
1.50
3.00
midazolam (μg/kg/inj)
Saline
Heroin 0.015 (mg/kg/inj)

C

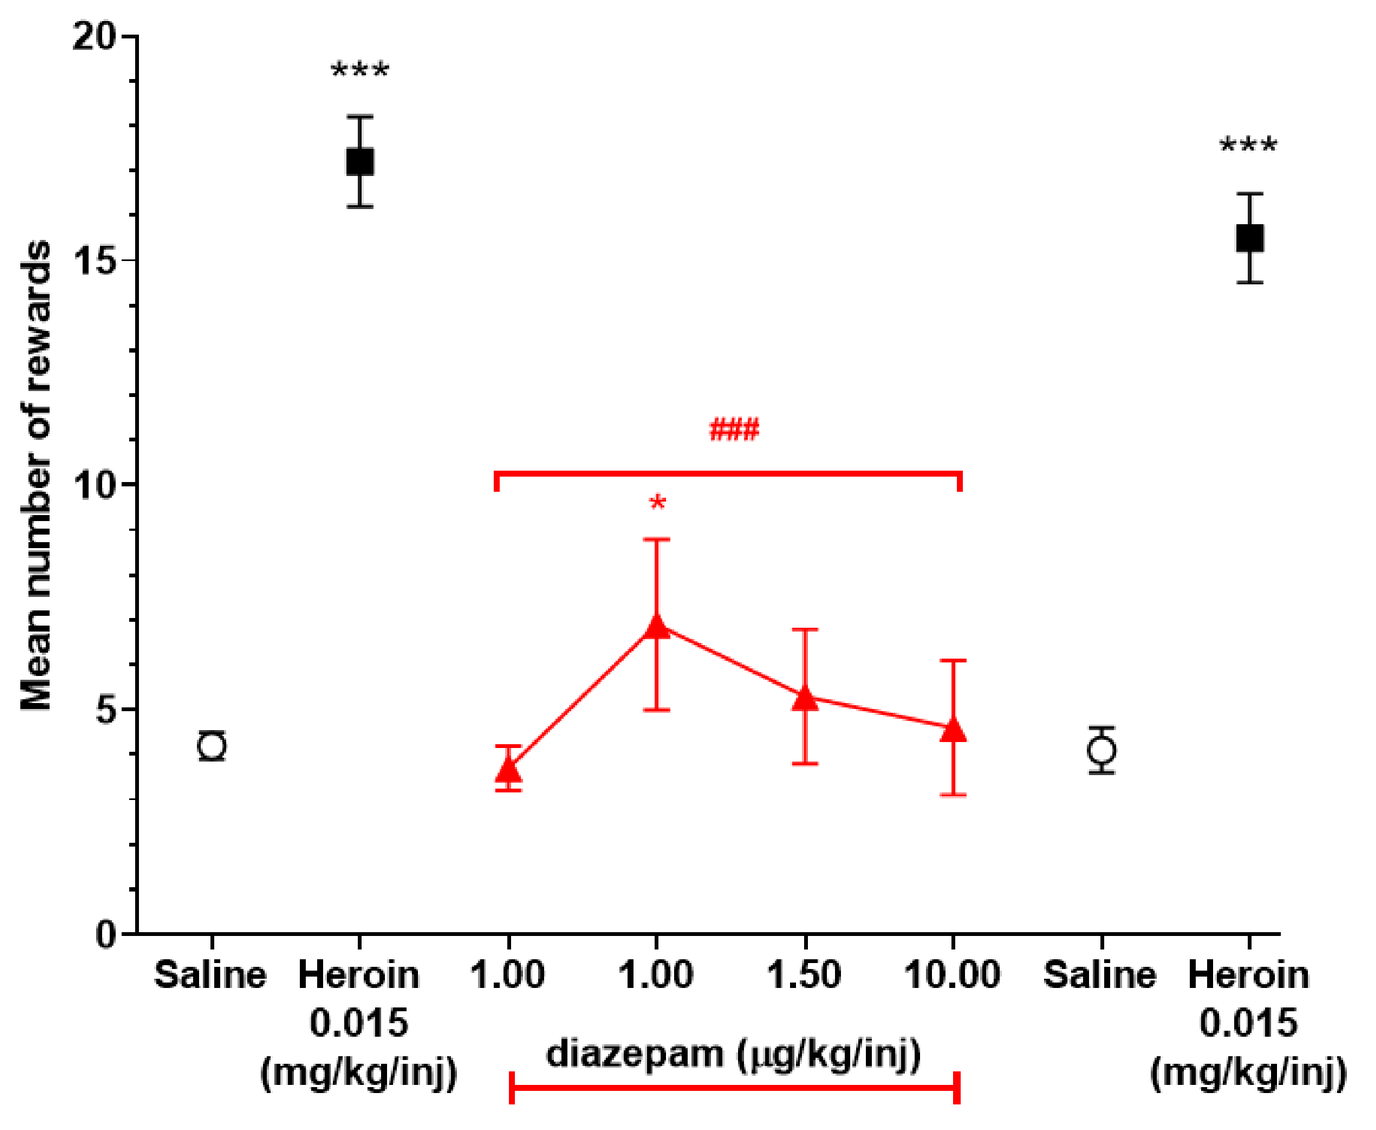
Mean number of rewards

###
*

Saline
Heroin 0.015 (mg/kg/inj)
1.00
1.00
1.50
10.00
diazepam (μg/kg/inj)
Saline
Heroin 0.015 (mg/kg/inj)

Effect of CBD (A) midazolam (B) and diazepam (C) in the heroin trained rat. Data are represented as the mean ± SEM number of injections per session during the last three sessions for each condition in rats responding under an FR3 schedule of *i.v.* self-administration. Means are back-transformed and adjusted for differences between animals. SEM are calculated from the residuals of the statistical model. Cannabidiol was compared to saline and heroin and family-wise error determined by Dunnett's test. Heroin was compared to saline by the multiple t test. Significantly different to saline *p<0.05, ***p<0.001. Significantly different to heroin ###P<0.001.

- Heroin maintained self-administration in rats at levels significantly greater than saline (17.6 ± 0.5 infusions/session, n = 39 vs 3.7 ± 0.2 infusions/session, n = 39; p<0.001).
- CBD (at 100 but not 10 or 300 μg/kg/infusion), maintained a low level of self-administration [6.9±1.8 infusions/session; n=8] at a level that was significantly greater than saline (p<0.05).
- The reference comparators, diazepam (1μg/kg/infusion) [7.0±2.1 infusions/session; n=8] and midazolam (1.5μg/kg/infusion) [7.3±1.3infusions/session; n=16] maintained significantly greater self-administration than saline (p<0.05), and met the criteria for weak positive reinforcers.

Evaluation of reinforcing effects of CBD in the midazolam-trained monkey

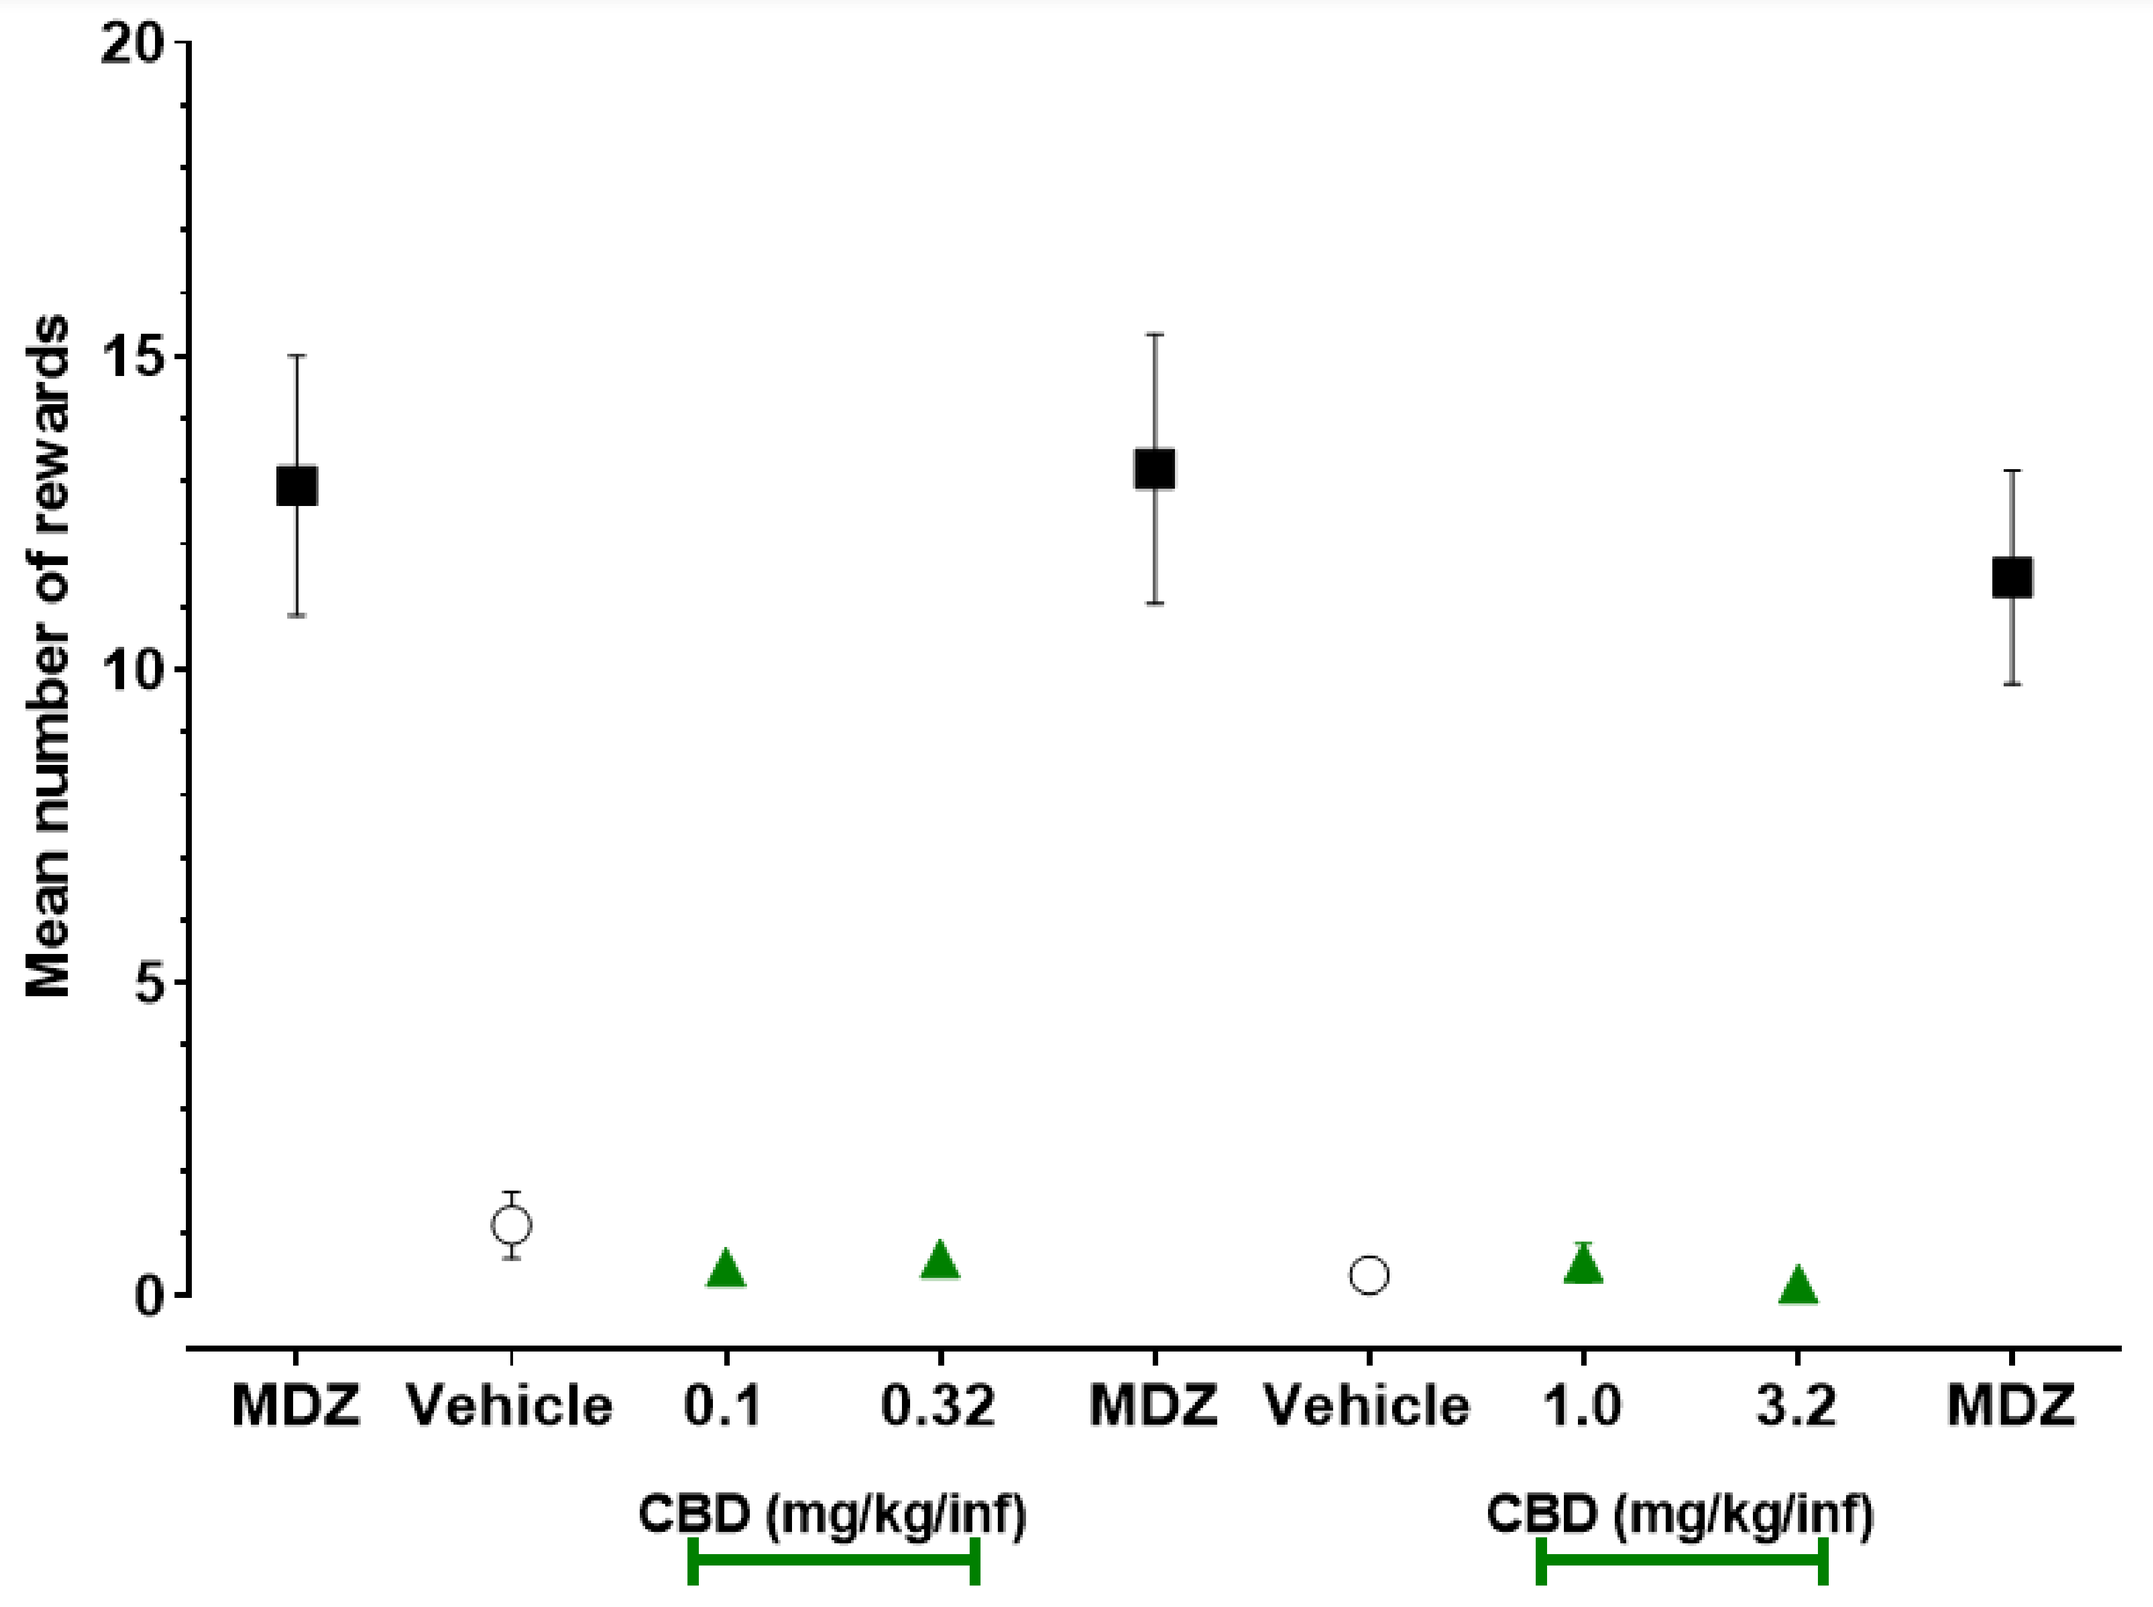
Mean number of rewards
20
15
10
5
0
MDZ
Vehicle
0.1
0.32
MDZ
Vehicle
1.0
3.2
MDZ
CBD (mg/kg/inf)
CBD (mg/kg/inf)

Data are represented as the mean ± SEM number of number of infusions received in the last three sessions of each condition; n=5. MDZ = midazolam. SEM for number of rewards in response to CBD and vehicle groups smaller than symbols.

- Monkeys received a mean of 12.9 ± 2.1 infusions per session of midazolam and 1.1 ± 0.5 infusions per session of vehicle.
- Responding for CBD was not statistically different from vehicle, with the maximum number of infusions received per session varying from 0.2 ± 0.1 (3.2 mg/kg/infusion) to 0.60 ± 0.30 (0.32 mg/kg/infusion).
- When each of the three largest doses of CBD was given as a bolus *i.v.* injection, plasma C_{max} values were 478 ± 66, 1730 ± 173, and 5530 ± 421 ng/ml; these values were between 1.5 and 12-fold higher than those obtained in humans receiving CBD therapeutically.

METHODS

- Reinforcing effects of CBD were examined in ♂ and ♀ midazolam-trained monkeys self-administering 0.010 (n=3) or 0.032 (n=2) mg/kg/infusion of midazolam (*i.v.)* under a fixed-ratio schedule (FR30). When responding for midazolam was stable, midazolam was replaced with CBD vehicle (1:1:9 ethanol:emulphor:saline) or CBD (0.1, 0.32, 1.0, and 3.2 mg/kg/infusion *i.v.*). Vehicle and each dose of CBD was examined for ≥ four sessions until responding was stable.
- The reinforcing effects of CBD were also examined in ♂ heroin-trained Sprague-Dawley rats. Rats trained to stably self-administer heroin (15 μg/kg/inj.) on a fixed-ratio schedule (FR3) of reinforcement in 2-hr training sessions, underwent saline extinction followed by assessment of the reinforcing effects of CBD (20, 100, 500 μg/kg/infusion), diazepam (1, 3, 4.5 or 10 μg/kg/infusion), and midazolam (0.3, 1, 1.5, 2.25 or 3 μg/kg/infusion).
- The discriminative properties of CBD were evaluated in ♀ Lister hooded rats, trained to discriminate midazolam (0.5mg/kg *i.p.*) from saline in a 2-choice operant test. Lever-pressing was reinforced by sweetened milk rewards on a FR5 schedule. Having reached >75% correct responding for midazolam versus saline, CBD (20, 75, 150 mg/kg *p.o.*), midazolam (0.5, 1.0, 1.5 mg/kg *p.o.*), and alprazolam (0.125, 0.25, 0.5, 1.0 mg/kg *p.o.*) were examined.
- HPLC/MS/MS of plasma CBD exposure was performed for assessment of plasma [CBD].

Disclosures: These studies were sponsored by GW Research Ltd (Cambridge, UK). All authors met the ICMJE authorship criteria and had full access to relevant data. Neither honoraria nor payments were made for authorship. Roy Gray is an employee of GW Research Ltd. Epidiolex® is approved in the U.S. for seizures associated with Lennox-Gastaut syndrome or Dravet syndrome in patients ≥2 years of age.©2019 Greenwich Biosciences, Inc. All rights reserved.

Contact Information: medinfo@greenwichbiosciences.com

Presented: American College of Neuropsychopharmacology Annual Meeting; Orlando, FL; December 8–11, 2019.

To obtain a PDF of this poster:
- Scan the QR code or
- Visit www.gwqrcodes.com/207173

Charges may apply.
No personal information is stored.